663

Efficacy and Safety of Lefamulin Versus Moxifloxacin for *Legionella pneumophila* in Patients With Community-Acquired Bacterial Pneumonia: Pooled Results From the Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 Phase 3 Clinical Trials

Andrew F. Shorr,[1] Jennifer Schranz,[2] Lisa Goldberg,[2] Susanne Paukner,[3] Anita F. Das,[4] Gregory J. Moran,[5] Christian Sandrock,[6] Thomas M. File Jr,[7] Elizabeth Alexander,[2] Steven P. Gelone[2]

[1]Medstar Washington Hospital Center, Washington, DC, USA; [2]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA; [3]Nabriva Therapeutics GmbH, Vienna, Austria; [4]Das Consulting, Guerneville, CA, USA; [5]Olive View-UCLA Medical Center, Los Angeles, CA, USA; [6]UC Davis School of Medicine, Sacramento, CA, USA; [7]Summa Health, Akron, OH, USA

Presented by
Elizabeth Alexander, MD, MSc
Email: Elizabeth.Alexander@nabriva.com
Phone: 610-981-2840
Fax: 610-816-6639

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION & PURPOSE

- Pneumonia is associated with substantial morbidity, mortality, and economic burden and is among the leading causes of infection-related death in the United States[1,2]
- Among adults with pneumonia, approximately 14% of infections are caused by the atypical pathogens *Mycoplasma pneumoniae*, *Chlamydophila pneumoniae*, and *Legionella pneumophila*[3]
- Of the atypical pathogens, community-acquired bacterial pneumonia (CABP) caused by *L. pneumophila* is associated with the highest severity of disease and the quickest onset of illness[4]
 - Clinical features of pneumonia caused by *L. pneumophila* include fever with organ-specific symptoms and signs (eg, diarrhea or confusion) and fever with multisystem disease (including rhabdomyolysis with renal failure)[5]
 - Inadequate or delayed antimicrobial therapy has been shown to lead to increased mortality rates in patients with *L. pneumophila*,[6] and, compared with patients with pneumococcal pneumonia, patients with CABP caused by *L. pneumophila* are more likely to receive inappropriate empiric treatment and require admission to an intensive care unit[7]
 - Many patients with *L. pneumophila* are treated with macrolides or fluoroquinolones,[7] although increasing rates of resistance to these antibiotic classes[8] and safety concerns (eg, fluoroquinolone-associated disability)[9,10] have created a need for new treatment options
- Lefamulin (LEF), a first-in-class pleuromutilin antimicrobial approved for intravenous (IV) and oral use in adults with CABP,[11] inhibits protein synthesis and has demonstrated potent in vitro activity against typical (eg, *Streptococcus pneumoniae*, *Staphylococcus aureus*, *Haemophilus influenzae*) and atypical CABP pathogens, including those resistant to other major antibiotic classes[11-14]
 - In 2 in vitro studies of *L. pneumophila* isolates from Germany (*n*=30) and the United States (*n*=44), the minimum inhibitory concentrations at which 50%/90% of isolates were inhibited ($MIC_{50/90}$) with LEF were 0.06/0.5 µg/mL and 0.5/1 µg/mL, respectively
 - In 2 recent phase 3, double-blind, double-dummy clinical trials, treatment with LEF was noninferior to the standard of care, moxifloxacin (MOX), in adults with CABP[15,16]
- This investigation assessed the efficacy and safety of LEF vs MOX in adults with CABP caused by *L. pneumophila* using data from pooled analyses of the Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 phase 3 clinical trials

METHODS

Study Design

- Both studies were global, prospective, randomized, double-blind, double-dummy, phase 3 trials **(Figure 1)**[15,16]
- In LEAP 1, patients were randomized to receive LEF 150 mg IV every 12 hours (q12h) for 5–7 days or MOX 400 mg IV every 24 hours (q24h) for 7 days
 - Patients could switch to oral therapy (LEF 600 mg q12h or MOX 400 mg q24h) after 6 IV doses of study drug (~3 days) if predefined improvement criteria were met
- In LEAP 2, patients were randomized to receive oral LEF 600 mg q12h for 5 days or oral MOX 400 mg q24h for 7 days

Figure 1. LEAP 1 and LEAP 2 Study Design

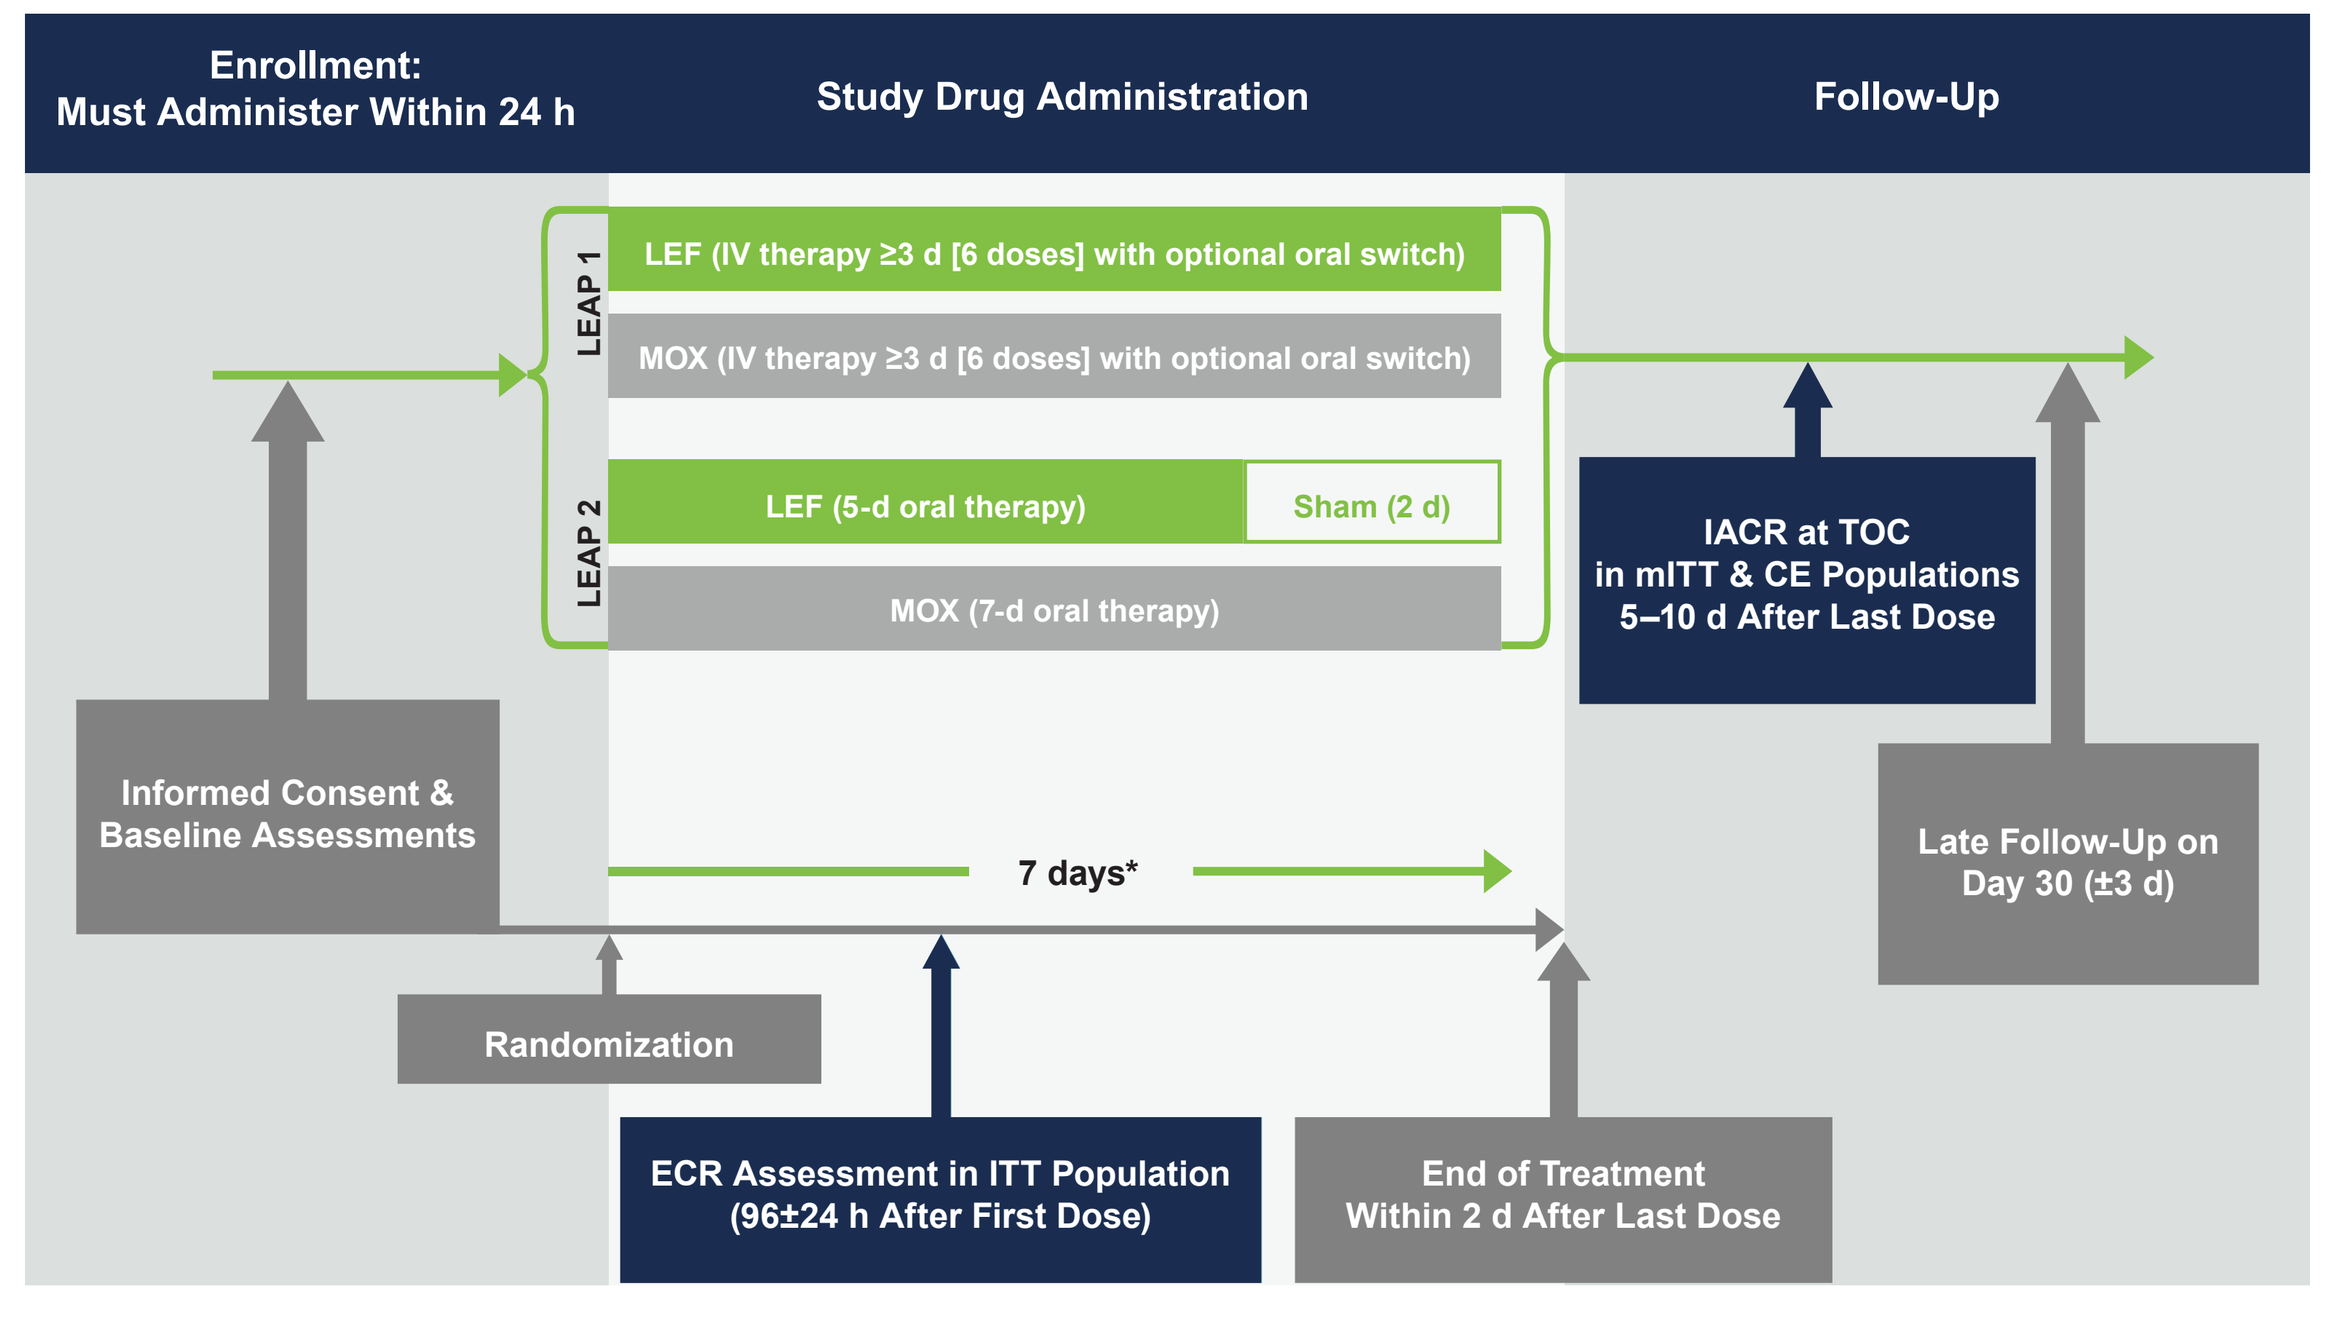

Enrollment:
Must Administer Within 24 h
Study Drug Administration
Follow-Up
LEAP 1
LEF (IV therapy ≥3 d [6 doses] with optional oral switch)
MOX (IV therapy ≥3 d [6 doses] with optional oral switch)
LEAP 2
LEF (5-d oral therapy)
Sham (2 d)
MOX (7-d oral therapy)
IACR at TOC
in mITT & CE Populations
5–10 d After Last Dose
Informed Consent &
Baseline Assessments
7 days*
Late Follow-Up on
Day 30 (±3 d)
Randomization
ECR Assessment in ITT Population
(96±24 h After First Dose)
End of Treatment
Within 2 d After Last Dose

CABP=community-acquired bacterial pneumonia; CE=clinically evaluable (patients who met predefined specified criteria related to protocol adherence); ECR=early clinical response (patient assessed as responder if alive, showed improvement in ≥2 CABP signs and symptoms, no worsening in any CABP sign or symptom, and no receipt of a concomitant nonstudy antibiotic for the current CABP episode); IACR=investigator assessment of clinical response (patients assessed as success if alive, with signs and symptoms of CABP resolved or improved such that no additional antibacterial therapy was administered for CABP); ITT=intent to treat (all randomized patients); IV=intravenous; LEAP=Lefamulin Evaluation Against Pneumonia; LEF=lefamulin; mITT=modified ITT (all randomized patients who received any amount of study drug); MOX=moxifloxacin; TOC=test-of-cure visit.
*In LEAP 1, the original protocol indicated a LEF treatment period of 5 days (but 10 days in patients with CABP due to *Legionella pneumophila* or methicillin-resistant *Staphylococcus aureus* [MRSA] or in patients with *Streptococcus pneumoniae* and bacteremia); however, this was later adjusted to 7 days (except in cases of confirmed MRSA, which continued to receive 10 days of treatment) to reduce medication errors and limit the burden on study sites.[15]

Patients and Assessments

- Adults with CABP of Pneumonia Outcomes Research Team (PORT) risk class III–V and II–IV were eligible for LEAP 1 and LEAP 2, respectively
- In both studies, the primary efficacy endpoint for the US Food and Drug Administration (FDA) was early clinical response (ECR) at 96±24 hours after first dose of study drug in the intent-to-treat (ITT) population
- The European Medicines Agency coprimary endpoints (FDA secondary endpoints) were investigator assessment of clinical response (IACR) at the test-of-cure (TOC) assessment 5–10 days after the last dose of study drug in the modified ITT and clinically evaluable populations

METHODS (continued)

- In both studies, baseline *L. pneumophila* was identified from specimens collected within 24 hours of the first dose of study drug by sputum culture, urine antigen testing (BinaxNOW®; Abbott Diagnostics, Santa Clara, CA, USA), serology (*L. pneumophila* group 1-6 indirect fluorescent antibody assay; Zeus Scientific, Branchburg, NJ, USA), or real-time polymerase chain reaction (RT-PCR) positive for the *ssrA* gene
 - Confirmatory identification and susceptibility testing of isolates, urine antigen testing, serology (≥4-fold increase in *L. pneumophila* antibody titer to ≥1:128), and RT-PCR were performed by a central laboratory and specialized laboratories (see Acknowledgments)
- Patients had to have baseline *L. pneumophila* to be included in the analyses described herein
 - Within this patient subgroup, efficacy analyses are presented for the microbiological intent-to-treat (microITT) population (randomized patients with ≥1 baseline CABP-causing pathogen), microITT-2 population (randomized patients with ≥1 baseline CABP-causing pathogen detected by a method other than PCR), and microbiologically evaluable (ME) population (met microITT and clinically evaluable population criteria)
 - Treatment-emergent adverse events (TEAEs) are presented for the microITT population

RESULTS

Patients and Baseline Characteristics

- The pooled phase 3 ITT population included 1289 patients (LEF, *n*=646; MOX, *n*=643)
- Within the overall pooled microITT population (LEF, *n*=364; MOX, *n*=345), *L. pneumophila* was identified in 9.3% of patients treated with LEF (34/364) and 9.0% of patients treated with MOX (31/345)
 - Patient demographics and baseline characteristics in this subgroup were generally similar to those of the overall ITT population, although more patients with *L. pneumophila* had PORT risk class IV pneumonia at baseline compared with the overall ITT population **(Table 1)**
- Serology and urinary antigen test were the 2 most common diagnostic modalities used to identify *L. pneumophila* at baseline **(Figure 2)**
 - Among patients with *L. pneumophila* at baseline, 31 (47.7%) patients had polymicrobial pneumonia; coinfection with a gram-positive pathogen (eg, *S. pneumoniae*) was recorded in 17 (26.2%) patients **(Figure 3)**
 - *L. pneumophila* isolates collected from sputum (*n*=2) displayed MIC values of 0.5–1 µg/mL for LEF and 0.03 µg/mL for MOX

Table 1. Demographics and Baseline Characteristics

Parameter	All Patients (Pooled ITT Population) LEF (*n*=646)	All Patients (Pooled ITT Population) MOX (*n*=643)	Patients With *L. pneumophila* (Pooled microITT Population) LEF (*n*=34)	Patients With *L. pneumophila* (Pooled microITT Population) MOX (*n*=31)
Age, y, median (range)	61 (19–97)	60 (19–93)	60 (25–89)	61 (26–89)
Male, *n* (%)	377 (58.4)	340 (52.9)	25 (73.5)	18 (58.1)
White, *n* (%)	513 (79.4)	509 (79.2)	31 (91.2)	28 (90.3)
PORT risk class, *n* (%)				
I/II*	184 (28.5)	192 (29.9)	7 (20.6)	7 (22.6)
III	341 (52.8)	334 (51.9)	19 (55.9)	16 (51.6)
IV/V*	121 (18.7)	117 (18.2)	8 (23.5)	8 (25.8)
Met minor ATS severity criteria,† *n* (%)	85 (13.2)	85 (13.2)	7 (20.6)	4 (12.9)
Met modified ATS severity criteria,‡ *n* (%)	53 (8.2)	57 (8.9)	5 (14.7)	4 (12.9)
Met SIRS criteria,§ *n* (%)	621 (96.1)	609 (94.7)	33 (97.1)	27 (87.1)
Multilobar pneumonia, *n* (%)	170 (26.3)	177 (27.5)	9 (26.5)	6 (19.4)
Bacteremic, *n* (%)	13 (2.0)	12 (1.9)	0	0
Renal status,‖ *n* (%)				
Normal	311 (48.1)	312 (48.5)	19 (55.9)	14 (45.2)
Mild impairment	201 (31.1)	192 (29.9)	12 (35.3)	10 (32.3)
Moderate impairment	125 (19.3)	132 (20.5)	3 (8.8)	7 (22.6)
Severe impairment	7 (1.1)	6 (0.9)	0	0
Prior antibiotic use,¶ *n* (%)	147 (22.8)	145 (22.6)	10 (29.4)	8 (25.8)

ATS=American Thoracic Society; BUN=blood urea nitrogen; CrCl=creatinine clearance; eCRF=electronic case report form; ITT=intent to treat; LEF=lefamulin; microITT=microbiological ITT; MOX=moxifloxacin; PORT=Pneumonia Outcomes Research Team; SIRS=systemic inflammatory response syndrome; WBC=white blood cell (count).
*PORT risk class I/II and IV/V for the pooled ITT population but PORT risk class II and IV, respectively, for patients with *Legionella pneumophila*. PORT risk class was calculated programmatically using data obtained at the site and reported in the eCRF and was not always consistent with the site-reported PORT risk class used for enrollment/stratification.
†Defined as presence of ≥3 of the following 9 criteria at baseline: respiratory rate ≥30 breaths/min, O_2 saturation <90% or PaO_2 <60 mm Hg, BUN ≥20 mg/dL, WBC <4000 cells/mm³, confusion, multilobar infiltrates, platelets <100,000 cells/mm³, temperature <36°C, or systolic blood pressure <90 mm Hg.[17]
‡Defined as presence of ≥3 of the following 6 criteria at baseline: respiratory rate ≥30 breaths/min, SpO_2/FiO_2 <274 where SpO_2/FiO_2 = 64+0.84 (PaO_2/FiO_2), BUN ≥20 mg/dL, confusion, age ≥65 years, or multilobar infiltrates.[18]
§Defined as having ≥2 of the following 4 criteria at baseline: temperature <36°C or >38°C; heart rate >90 bpm; respiratory rate >20 breaths/min; and WBC <4000 cells/mm³, WBC >12,000 cells/mm³, or immature polymorphonuclear neutrophils >10%.
‖National Kidney Foundation categories of renal impairment[19] based on baseline central laboratory serum creatinine. When baseline central laboratory serum creatinine was not available, local serum creatinine results were used. Renal impairment categories are: normal [CrCl ≥90 mL/min], mild [CrCl of 60 to <90 mL/min], moderate [CrCl of 30 to <60 mL/min], and severe [CrCl <30 mL/min].
¶Patients received a single dose of short-acting systemic antibacterial medication within 72 hours before randomization; randomization was stratified and capped such that no more than 25% of the total ITT population met these criteria.

RESULTS (continued)

Figure 2. Diagnostic Modalities for Patients With *L. pneumophila* at Baseline* (Combined Treatment Groups)

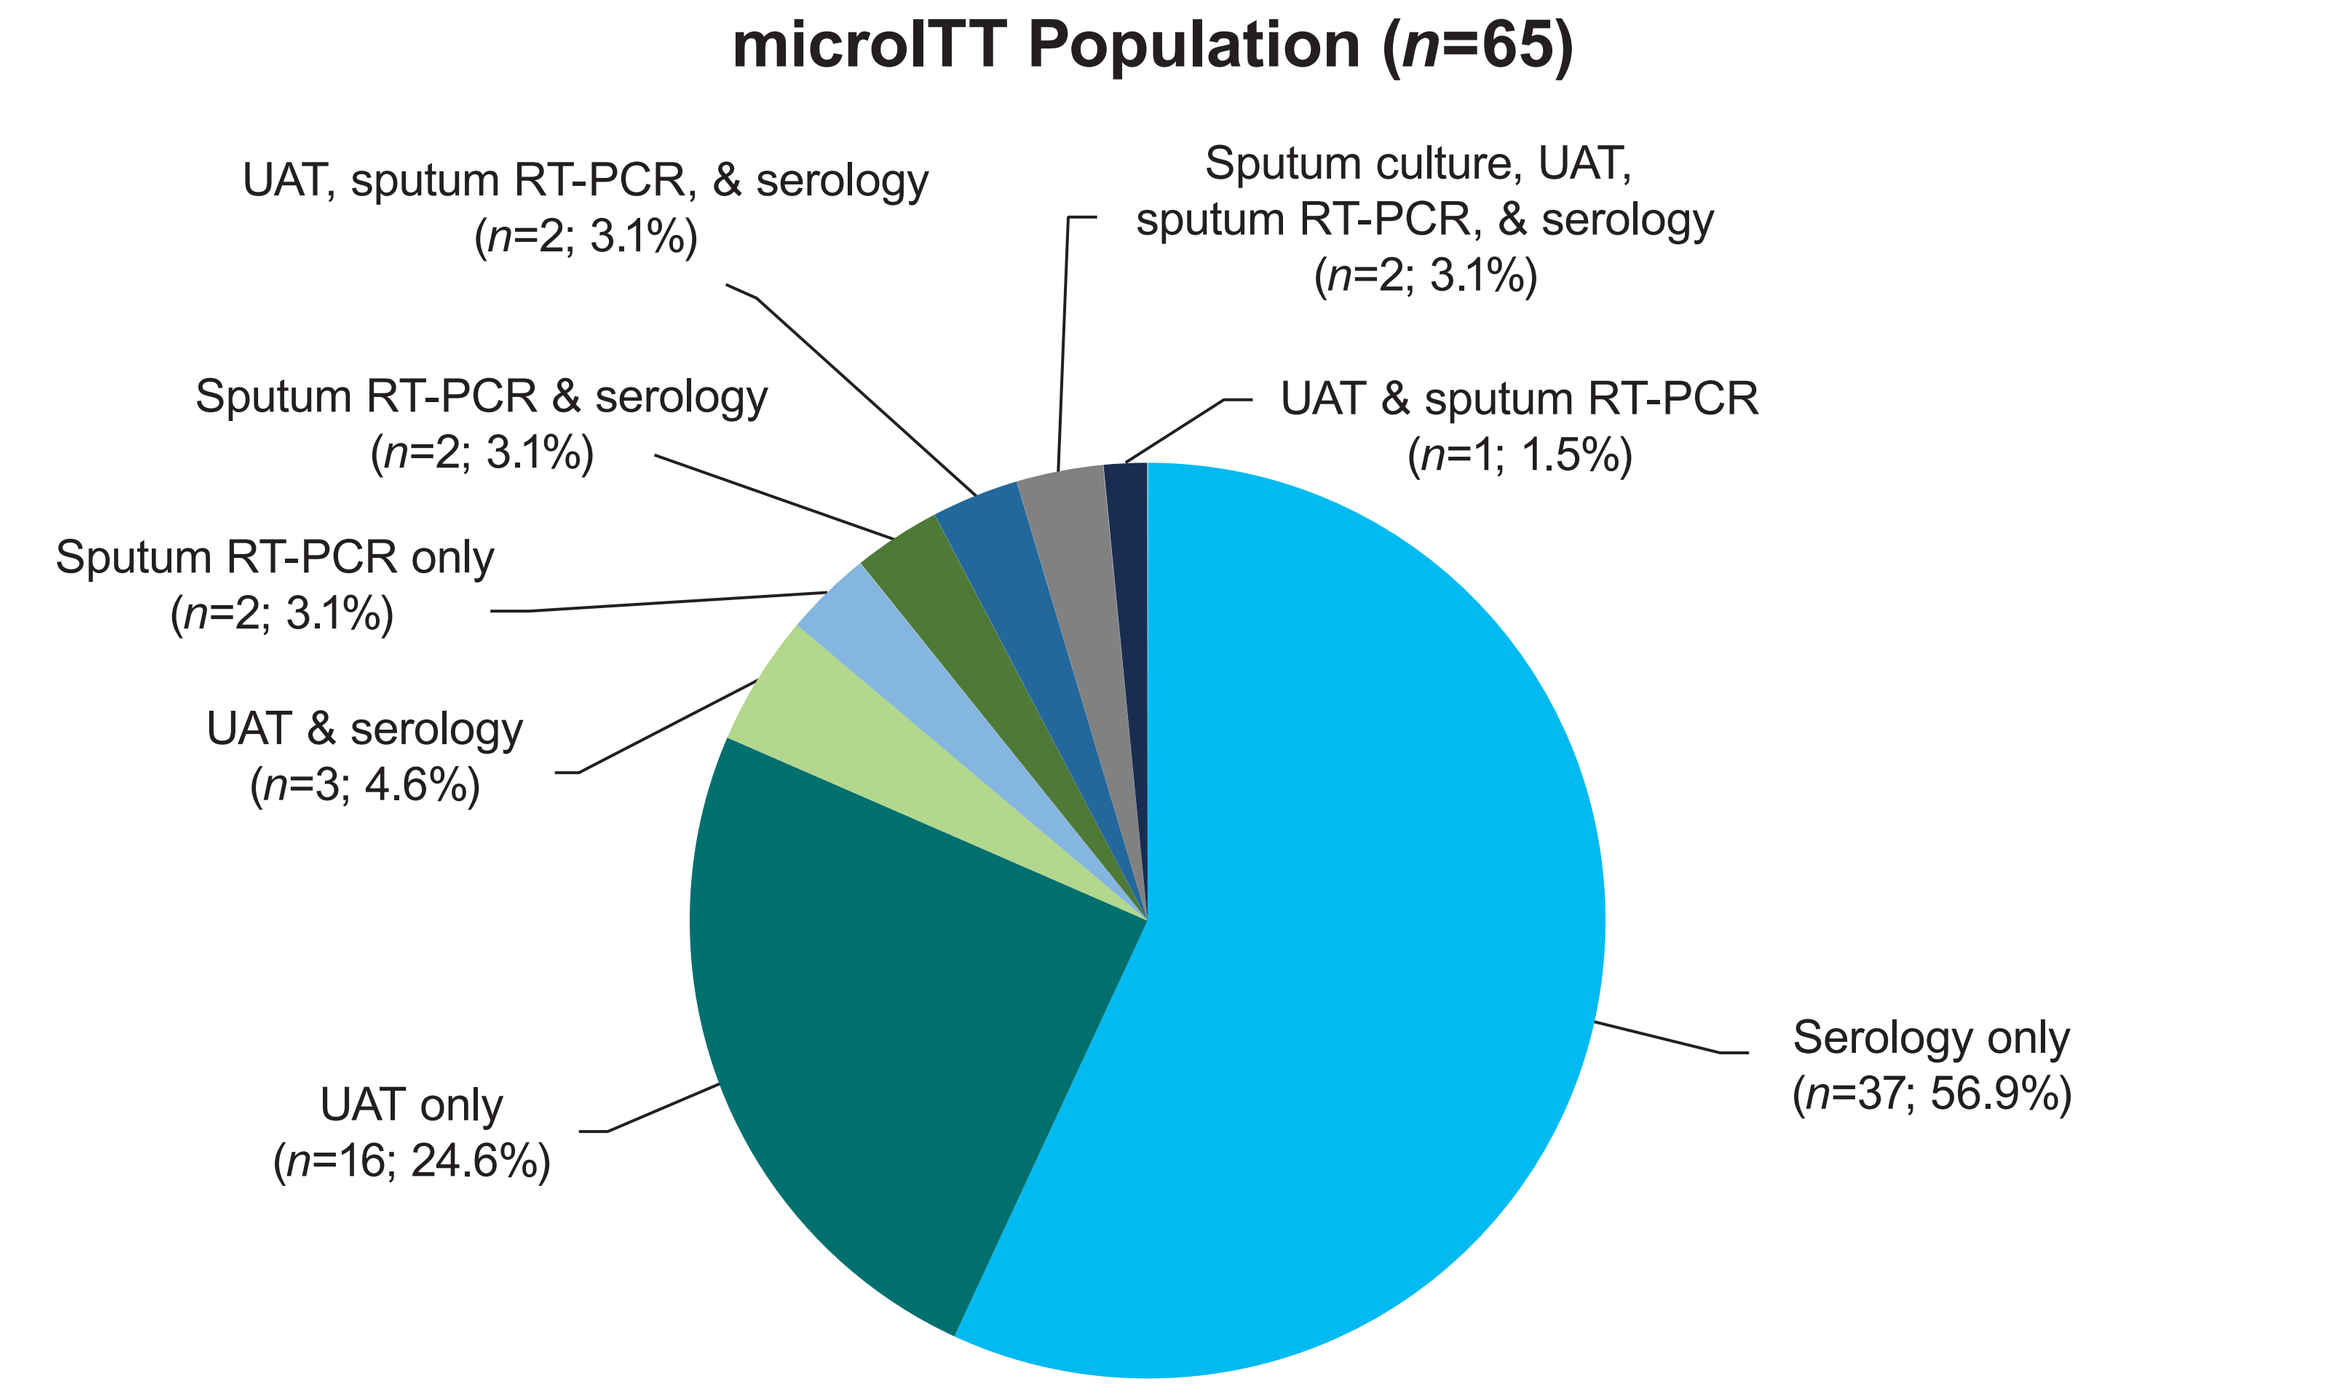

microITT Population (n=65)
UAT, sputum RT-PCR, & serology
(n=2; 3.1%)
Sputum culture, UAT,
sputum RT-PCR, & serology
(n=2; 3.1%)
Sputum RT-PCR & serology
(n=2; 3.1%)
UAT & sputum RT-PCR
(n=1; 1.5%)
Sputum RT-PCR only
(n=2; 3.1%)
UAT & serology
(n=3; 4.6%)
UAT only
(n=16; 24.6%)
Serology only
(n=37; 56.9%)

microITT=microbiological intent to treat; *n*=number of patients with *Legionella pneumophila* at baseline; RT-PCR=real-time polymerase chain reaction; UAT=urine antigen testing.
*A patient could have had >1 pathogen identified. Multiple isolates of the same species from the same patient identified by the same testing modality were counted only once. Patients were only counted once for each pathogen based on the unique diagnostic modality or combination of diagnostic modalities by which the pathogen was identified. Qualification of *L. pneumophila* as a baseline pathogen via serology testing required a ≥4-fold increase in *L. pneumophila* antibody titer to ≥1:128 between the baseline and convalescent samples. In sputum RT-PCR, the sample had to test positive for the *ssrA* gene.

Figure 3. Pathogen Distribution for Patients With *L. pneumophila* at Baseline* (Pooled microITT Population [Combined Treatment Groups])

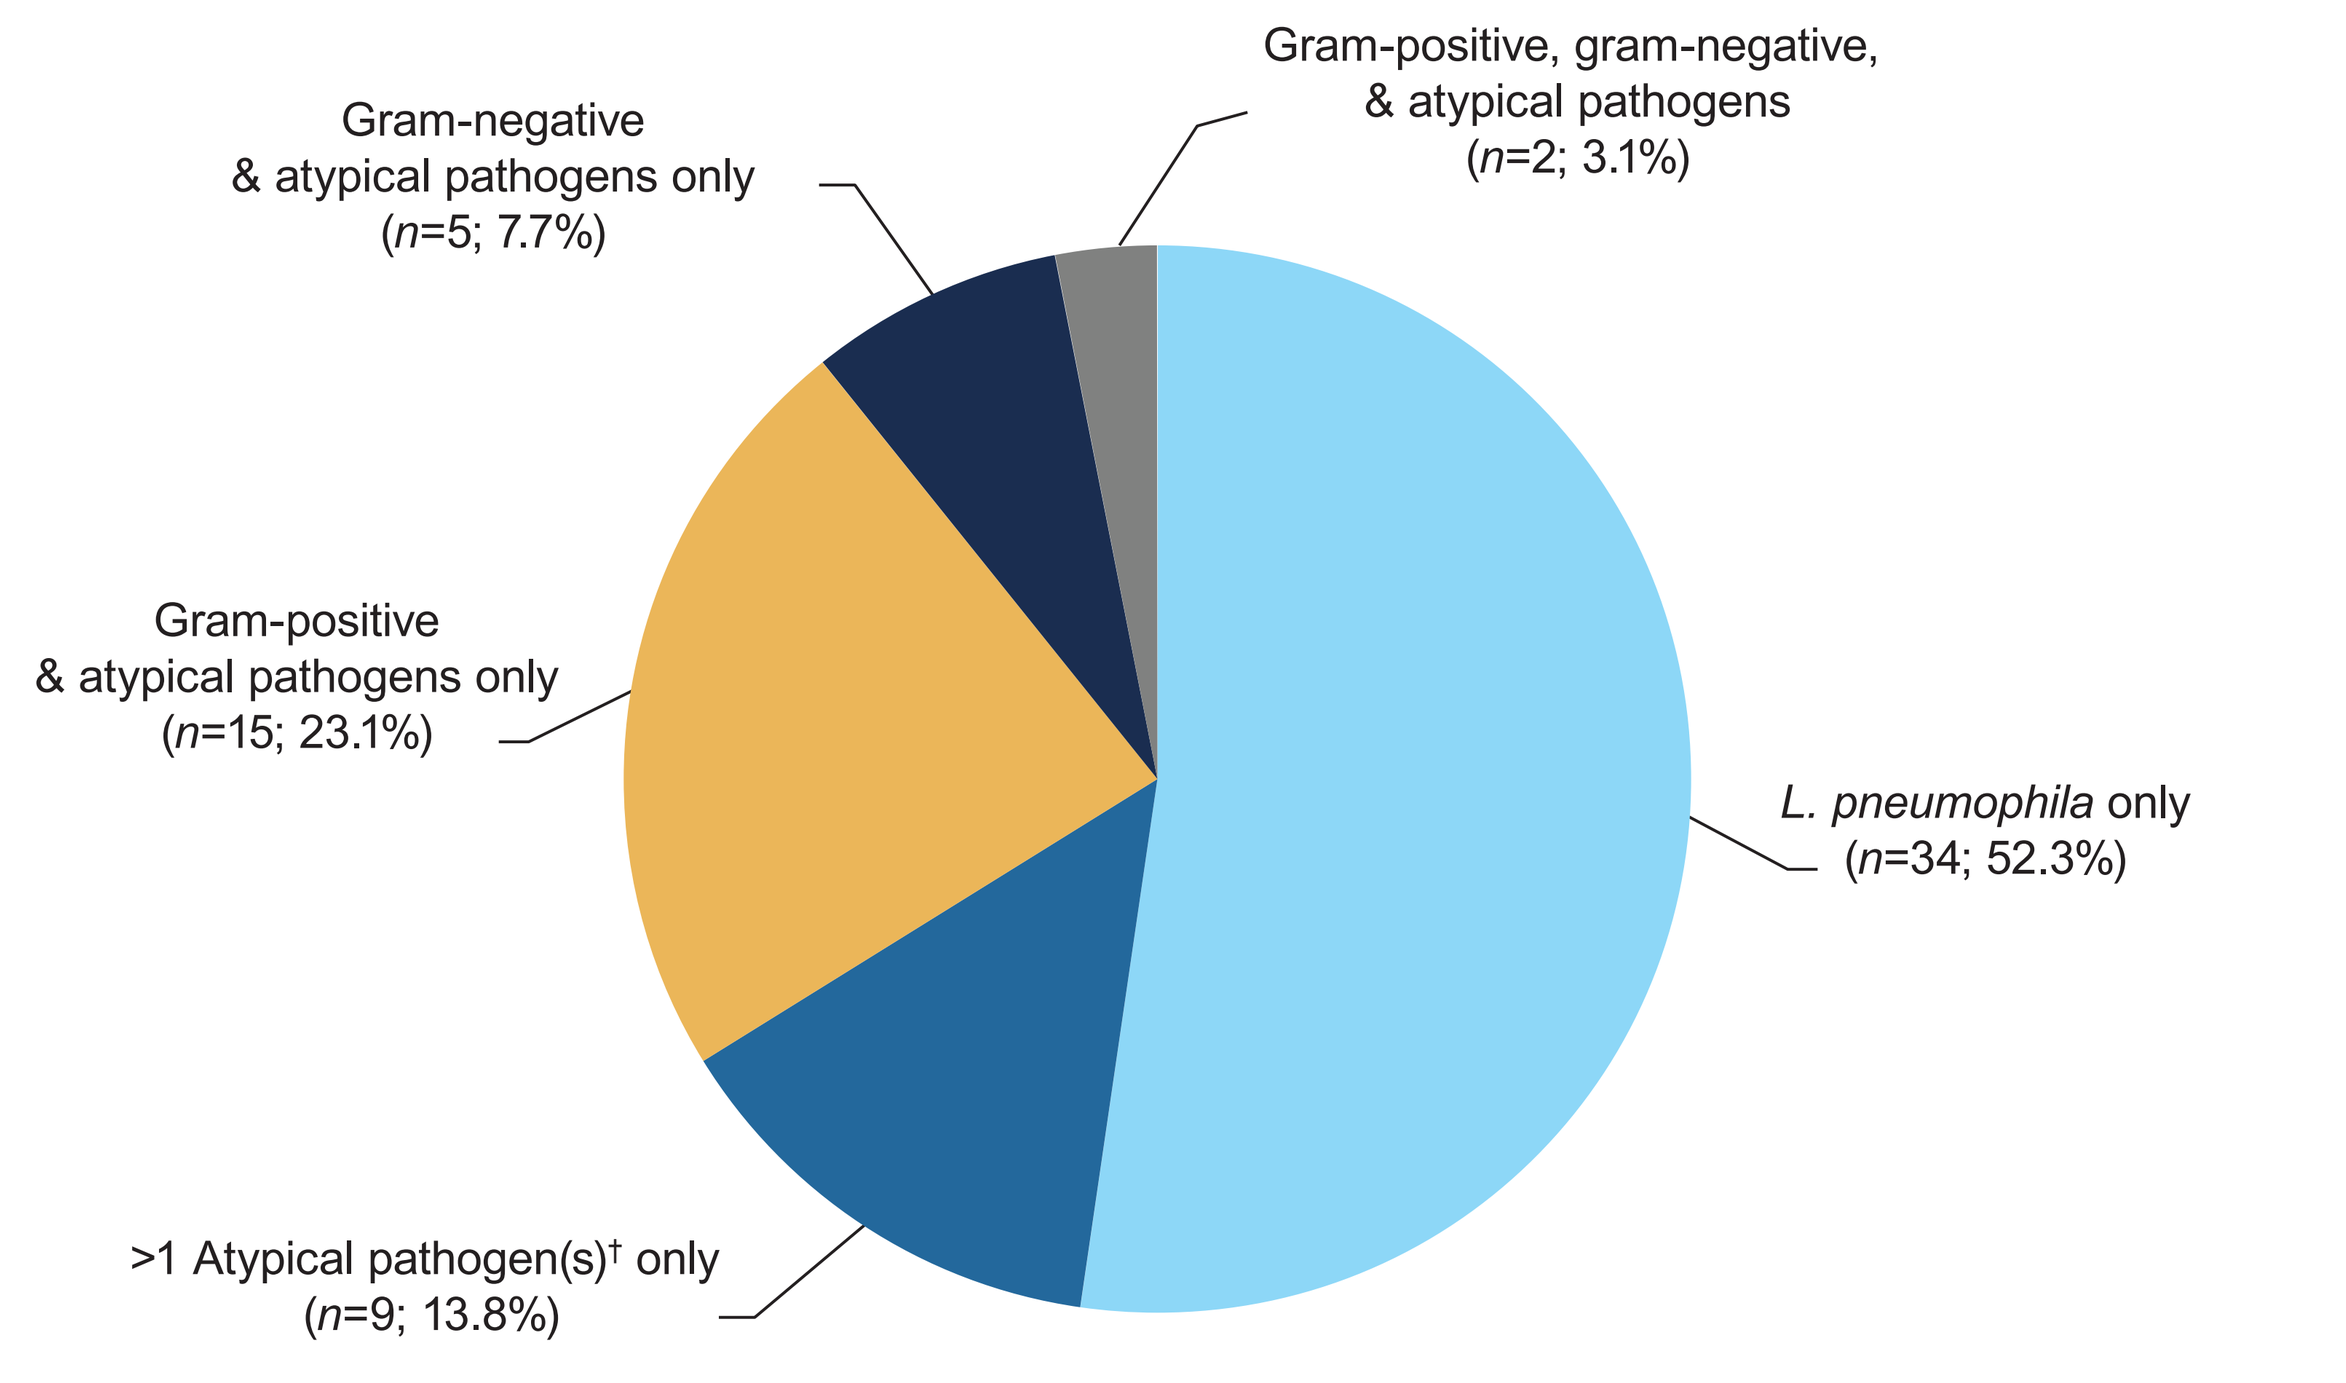

Gram-negative
& atypical pathogens only
(n=5; 7.7%)
Gram-positive, gram-negative,
& atypical pathogens
(n=2; 3.1%)
Gram-positive
& atypical pathogens only
(n=15; 23.1%)
L. pneumophila only
(n=34; 52.3%)
>1 Atypical pathogen(s)† only
(n=9; 13.8%)

microITT=microbiological intent to treat; *n*=number of patients with *Legionella pneumophila* at baseline.
*A patient could have had >1 pathogen identified. Patients were only counted once based on their unique pathogen grouping.
†Atypical pathogens included *L. pneumophila*, *Chlamydophila pneumoniae*, and *Mycoplasma pneumoniae*.

Efficacy

- Among patients with *L. pneumophila* at baseline, ECR rates in the microITT and microITT-2 populations were high and similar between treatment groups **(Figure 4)**
 - These results were consistent with ECR rates in the overall pooled microITT (LEF, 89.3%; MOX, 93.0%; difference, –3.7; 95% CI, –7.9 to 0.5) and microITT-2 (LEF, 90.0%; MOX, 92.8%; difference, –3.1; 95% CI, –8.7 to 2.6) populations
- Patients with *L. pneumophila* at baseline achieved high IACR success rates at TOC in the microITT, microITT-2, and ME populations that were similar between treatment groups **(Figure 4A)** and consistent with findings observed in the overall pooled populations
- Microbiological response of success at TOC in the microITT population, which relied on clinical responses in most instances as follow-up cultures were not performed, was comparable between treatment groups in patients with *L. pneumophila* at baseline; findings in the microITT-2 and ME populations were consistent **(Figure 4B)**

Figure 4. (A) Early Clinical Response and Investigator Assessment of Clinical Response at TOC and (B) Microbiological Response of Success* at TOC by Analysis Population in Patients With *L. pneumophila* at Baseline

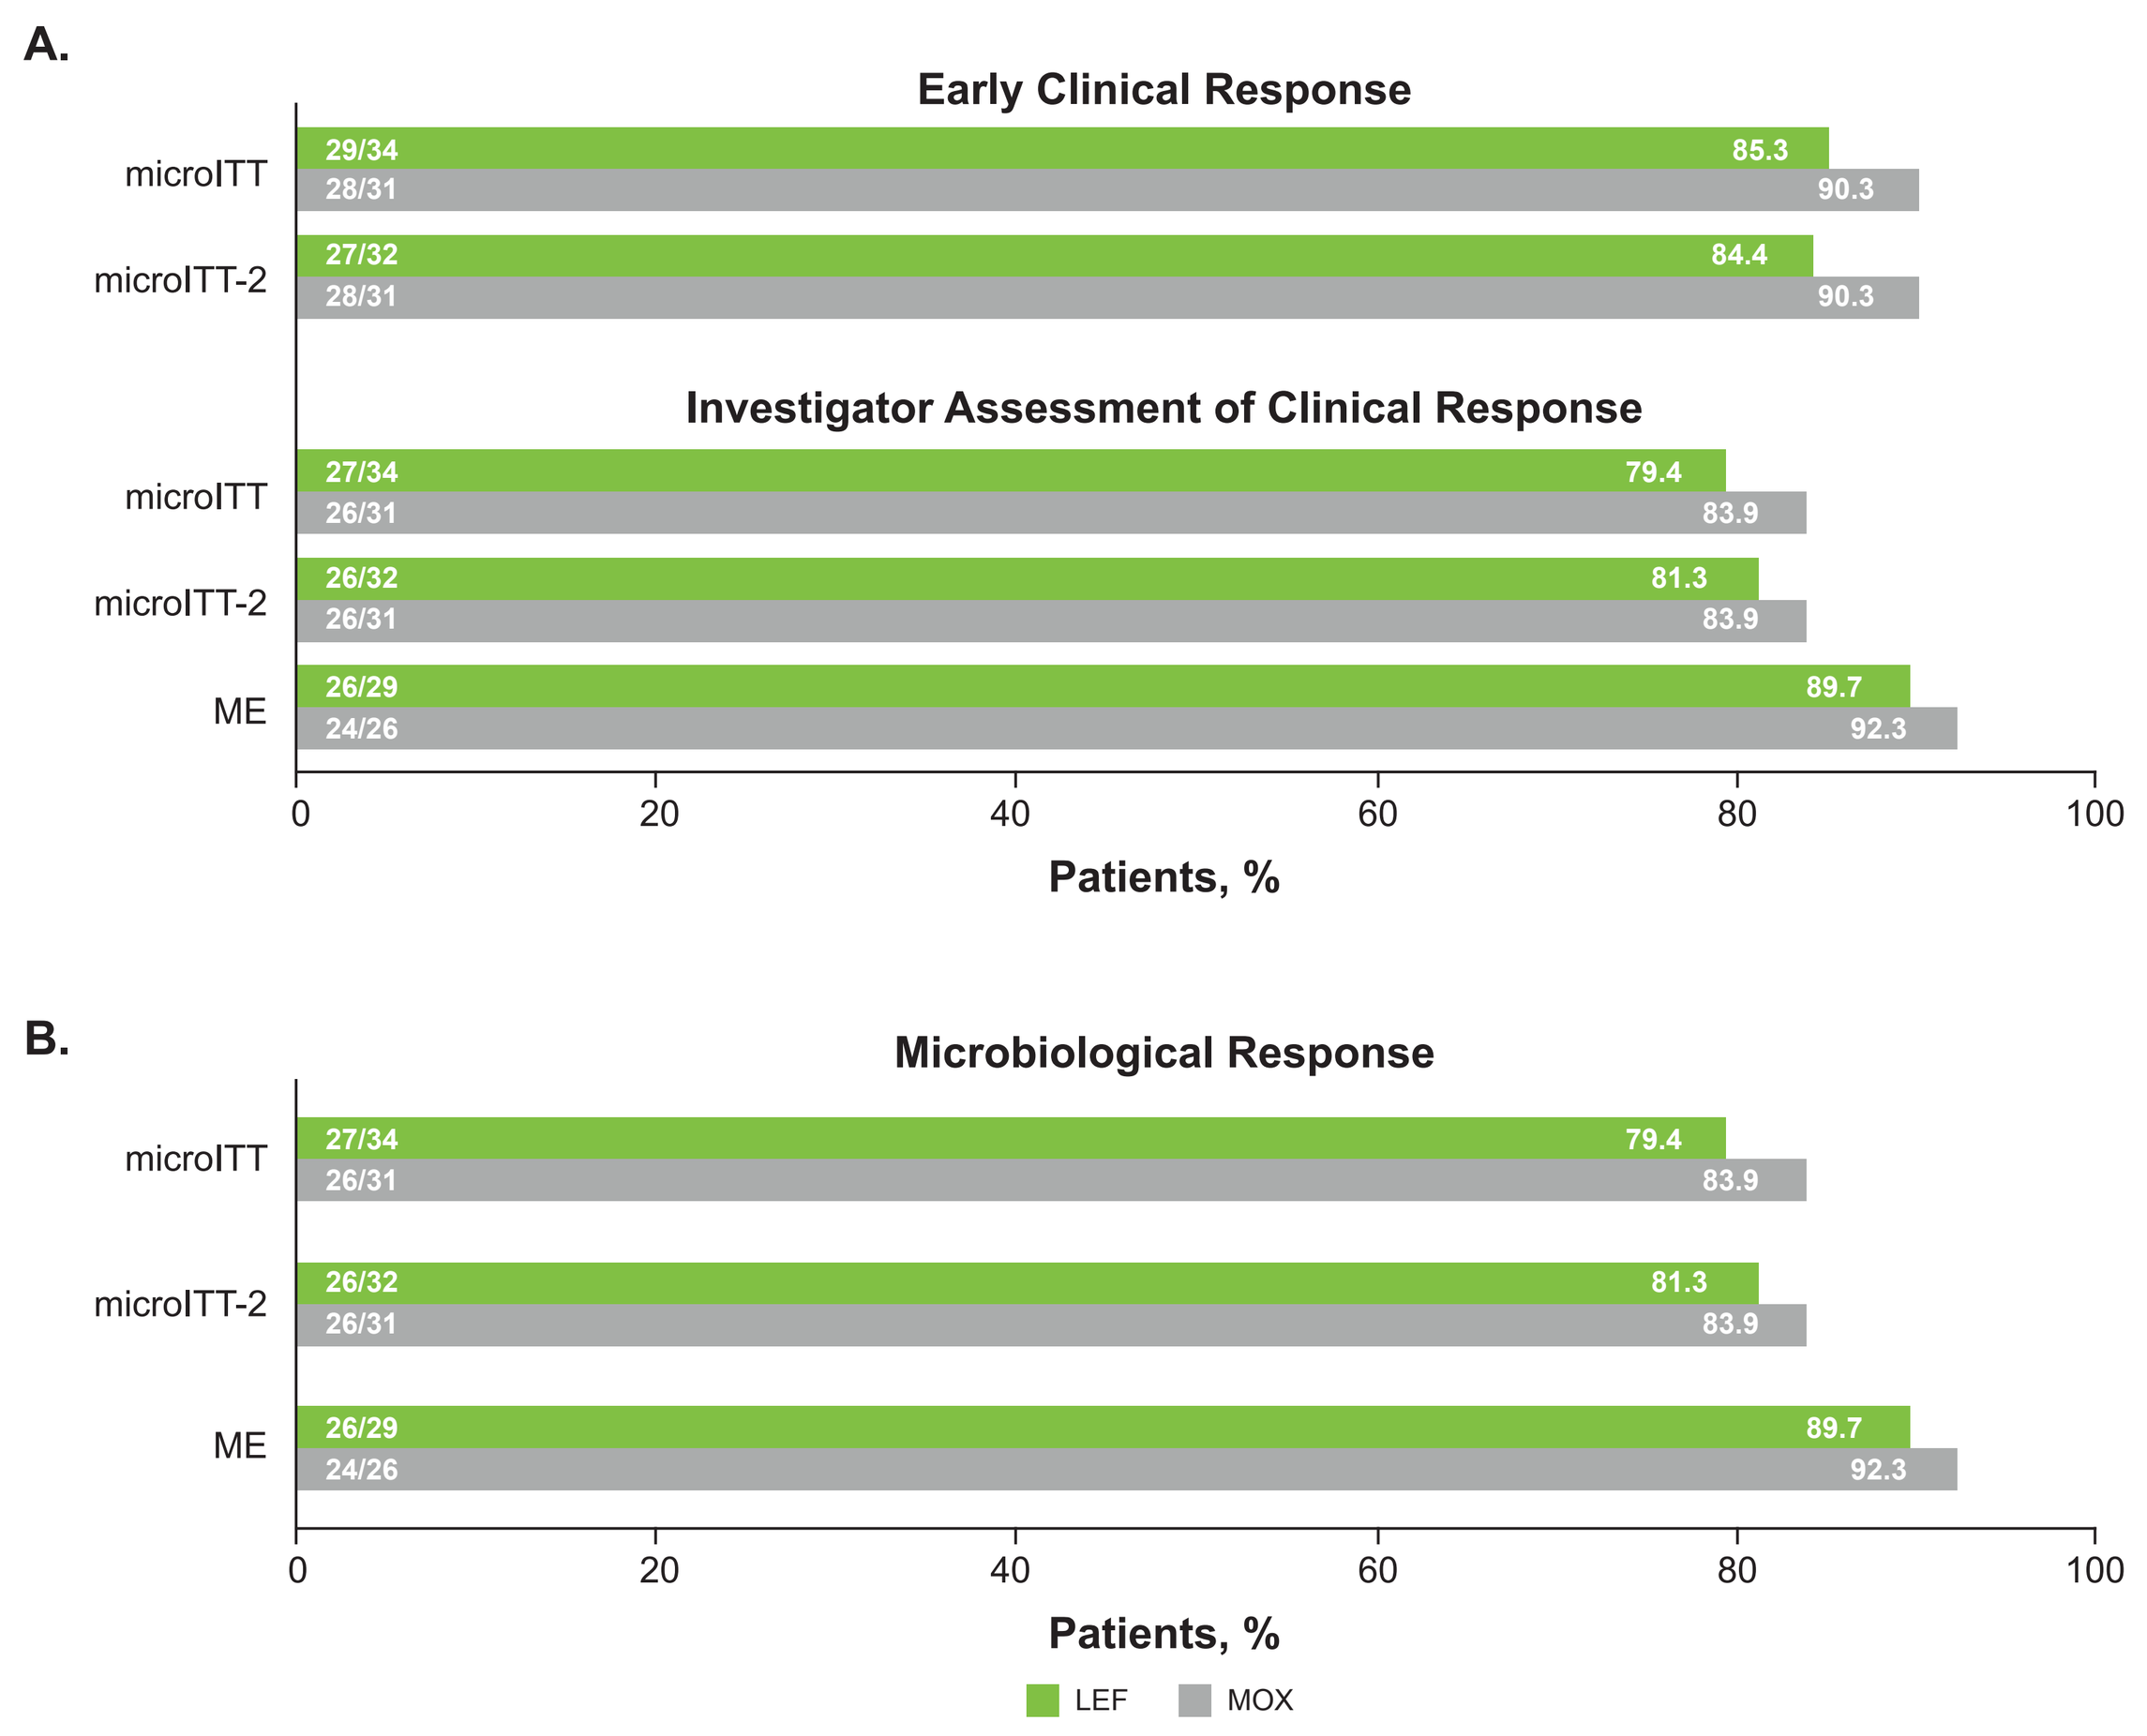

A.
Early Clinical Response
microITT
29/34
85.3
28/31
90.3
microITT-2
27/32
84.4
28/31
90.3
Investigator Assessment of Clinical Response
microITT
27/34
79.4
26/31
83.9
microITT-2
26/32
81.3
26/31
83.9
ME
26/29
89.7
24/26
92.3
0
20
40
60
80
100
Patients, %
B.
Microbiological Response
microITT
27/34
79.4
26/31
83.9
microITT-2
26/32
81.3
26/31
83.9
ME
26/29
89.7
24/26
92.3
0
20
40
60
80
100
Patients, %
LEF
MOX

EOT=end of treatment; IACR=investigator assessment of clinical response; LEF=lefamulin; ME=microbiologically evaluable; microITT=microbiological intent to treat; MOX=moxifloxacin; TOC=test-of-cure visit.
*Microbiological response of success at TOC was defined as either microbiologic eradication (absence of the baseline causative pathogen from repeat cultures obtained between EOT and TOC) or presumed eradication (the IACR at TOC was success, and culture was not repeated at TOC).

Safety

- Among patients with *L. pneumophila* at baseline, TEAE rates were 32.4% (11/34) and 32.3% (10/31) in the LEF and MOX groups, respectively **(Table 2)**
 - Most TEAEs were mild or moderate in severity and rarely led to study drug discontinuation
 - All serious TEAEs were unrelated to treatment and no deaths occurred
 - Results were consistent with those observed in the overall pooled safety population
- The system organ class with the most TEAEs among patients with *L. pneumophila* at baseline was 'respiratory, thoracic, and mediastinal disorders' **(Table 3)**
 - The only TEAEs reported by more than 1 patient were increased white blood cell count (LEF, 0; MOX, 2 [6.5%]), decreased lymphocyte count (LEF, 1 [2.9%]; MOX, 1 [3.2%]), and pulmonary embolism (LEF, 1 [2.9%]; MOX, 1 [3.2%])

Table 2. Overall Summary of TEAEs

Patients, *n* (%)	All Patients (Pooled Safety Population) LEF (*n*=641)	All Patients (Pooled Safety Population) MOX (*n*=641)	Patients With *L. pneumophila* (Pooled microITT Population) LEF (*n*=34)	Patients With *L. pneumophila* (Pooled microITT Population) MOX (*n*=31)
Any TEAE	224 (34.9)	195 (30.4)	11 (32.4)	10 (32.3)
Mild	119 (18.6)	117 (18.3)	5 (14.7)	4 (12.9)
Moderate	78 (12.2)	55 (8.6)	5 (14.7)	3 (9.7)
Severe	27 (4.2)	23 (3.6)	1 (2.9)	3 (9.7)
Related TEAE	99 (15.4)	68 (10.6)	1 (2.9)	2 (6.5)
Serious TEAE	36 (5.6)	31 (4.8)	3 (8.8)	2 (6.5)
Related serious TEAE	3 (0.5)	2 (0.3)	0	0
TEAE leading to death	11 (1.7)	8 (1.2)	0	0
28-d all-cause mortality – deceased at Day 28*	8 (1.2)	7 (1.1)	0	0
TEAE leading to study drug discontinuation	20 (3.1)	21 (3.3)	0	2 (6.5)

ITT=intent to treat; LEF=lefamulin; microITT=microbiological ITT; MOX=moxifloxacin; TEAE=treatment-emergent adverse event.
*Assessed in the ITT population (LEF, *n*=646; MOX, *n*=643).

Table 3. TEAE System Organ Classes Reported by ≥3 Patients With *L. pneumophila* at Baseline

System Organ Class* Patients, *n* (%)	All Patients (Pooled Safety Population) LEF (*n*=641)	All Patients (Pooled Safety Population) MOX (*n*=641)	Patients With *L. pneumophila* (Pooled microITT Population) LEF (*n*=34)	Patients With *L. pneumophila* (Pooled microITT Population) MOX (*n*=31)
Respiratory, thoracic, and mediastinal disorders	29 (4.5)	28 (4.4)	4 (11.8)	1 (3.2)
Blood and lymphatic system disorders	9 (1.4)	9 (1.4)	2 (5.9)	1 (3.2)
Infections and infestations	47 (7.3)	40 (6.2)	2 (5.9)	1 (3.2)
Gastrointestinal disorders	84 (13.1)	65 (10.1)	1 (2.9)	2 (6.5)
Investigations	31 (4.8)	26 (4.1)	1 (2.9)	2 (6.5)

LEF=lefamulin; microITT=microbiological intent to treat; MOX=moxifloxacin; PT=preferred term; SOC=system organ class; TEAE=treatment-emergent adverse event.
*Although a patient may have had >1 TEAE, the patient was counted only once within an SOC category and once within a PT category. The same patient may have contributed ≥2 PTs in the same SOC category, but the patient was only counted once towards that SOC category.

CONCLUSIONS

- Baseline clinical characteristics of patients with *L. pneumophila* were similar to those of the general patient population with CABP,[20] although a slightly higher disease severity (ie, more PORT risk class IV patients) was noted
- Therapy with LEF led to high efficacy rates (ECR, IACR success, and microbiological response of success) in patients with CABP infected with *L. pneumophila*, including when LEF was given as short-course (5-day) oral therapy
- The safety profile with LEF was similar between the overall population and patients with *L. pneumophila*; most TEAEs were mild to moderate in severity, and no deaths occurred in either treatment group
- LEF may provide a new empiric IV and oral monotherapy alternative to fluoroquinolones and macrolides in patients with CABP infected with *L. pneumophila*

REFERENCES

(1) McDermott KW, et al. Trends in hospital inpatient stays in the United States, 2005-2014: statistical brief #225. *Healthcare Cost and Utilization Project (HCUP) Statistical Briefs*. Rockville, MD: Agency for Healthcare Research and Quality; 2017.
(2) Xu J, et al. Deaths: final data for 2016. US Department of Health and Human Services, Centers for Disease Control and Prevention, National Center for Health Statistics. Available at: https://www.cdc.gov/nchs/data/nvsr/nvsr67/nvsr67_05.pdf. Accessed August 26, 2019.
(3) Marchello C, et al. *Ann Fam Med.* 2016;14(6):552-566.
(4) Arnold FW, et al. *Semin Respir Crit Care Med.* 2016;37(6):819-828.
(5) Phin N, et al. *Lancet Infect Dis.* 2014;14(10):1011-1021.
(6) Sharma L, et al. *Clin Chest Med.* 2017;38(1):45-58.
(7) Viasus D, et al. *Medicine (Baltimore).* 2013;92(1):51-60.
(8) Peyrani P, et al. *Expert Rev Respir Med.* 2019;13(2):139-152.
(9) US Food and Drug Administration. FDA drug safety communication: FDA updates warnings for oral and injectable fluoroquinolone antibiotics due to disabling side effects. Available at: https://www.fda.gov/Drugs/DrugSafety/ucm511530.htm. Accessed August 26, 2019.
(10) US Food and Drug Administration. FDA drug safety communication: FDA reinforces safety information about serious low blood sugar levels and mental health side effects with fluoroquinolone antibiotics; requires label changes. Available at: https://www.fda.gov/downloads/Drugs/DrugSafety/UCM612834.pdf. Accessed June 23, 2019.
(11) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(12) Waites KB, et al. *Antimicrob Agents Chemother.* 2017;61(2):e02008-16.
(13) Paukner S, et al. *Antimicrob Agents Chemother.* 2019;63(4):e02161-18.
(14) Sader HS, et al. *J Antimicrob Chemother.* 2012;67(5):1170-1175.
(15) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(16) Alexander E, et al. *JAMA.* 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].
(17) Mandell LA, et al. *Clin Infect Dis.* 2007;44(suppl 2):S27-S72.
(18) Li HY, et al. *Medicine (Baltimore).* 2015;94(36):e1474.
(19) Cockcroft DW and Gault MH. *Nephron.* 1976;16(1):31-41.
(20) Kolditz M, et al. *Thorax.* 2015;70(6):551-558.

Acknowledgments

We thank the personnel of Covance Central Laboratory Services (Indianapolis, IN, USA), UAB Diagnostic Mycoplasma Laboratory (Birmingham, AL, USA), Special Pathogens Laboratory-The Legionella Experts® (Pittsburgh, PA, USA), Research Unit on Respiratory Pathogens at RSPH (Emory University, Atlanta, GA, USA), and Accelero Bioanalytics (Berlin, Germany) for assistance in the performance of confirmatory and specialized testing.

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Andrew F. Shorr has served as a consultant for or received grant support from Achaogen, Melinta, Merck, Nabriva Therapeutics, Paratek, and Tetraphase. Jennifer Schranz, Lisa Goldberg, Susanne Paukner, Elizabeth Alexander, and Steven P. Gelone are employees of/stockholders in Nabriva Therapeutics plc. Anita F. Das has served as a consultant for Achaogen, AntibioTx, Boston Pharmaceuticals, Cempra, ContraFect, InterumTx, Nabriva Therapeutics, Paratek, Tetraphase, Theravance, UTILITY, Wockhardt, and Zavante. Gregory J. Moran has received grants from ContraFect and Nabriva Therapeutics. Christian Sandrock has served as a consultant for Allergan and Nabriva Therapeutics, has received grants from the National Institutes of Health and the Health Resources & Services Administration, and has received nonfinancial support from the State of California. Thomas M. File Jr. has served as a consultant for bioMérieux, Curetis, Melinta, Merck, Motif BioSciences, Nabriva Therapeutics, Paratek, Pfizer, and Shinogi Inc., and has received grants from Nabriva Therapeutics.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_LEAP1_2_Legionella

IDWeek 2019: October 2–6, Washington, DC, USA